ON101 SIGNIFICANTLY ENHANCES HEALING AND REDUCES AMPUTATION RATES IN INFECTED DIABETIC FOOT ULCERS (DFUs)

Shun-Cheng Chang, MD
Division of Plastic Surgery, Integrated Burn & Wound Care Center, Department of Surgery, Shuang-Ho Hospital, New Taipei City, Taiwan

Introduction

Infection is a leading contributor to high amputation rates among patients with diabetic foot ulcers(DFUs). Standard care for infected DFUs typically includes systemic antibiotics and silver-impregnated dressings, but clinical outcomes remain suboptimal, with published wound closure rates of only 27.5% at 6 months and 44.5% at 12 months. Modulating macrophage-driven immune responses within the infected wound microenvironment has emerged as a promising strategy to enhance healing. This study evaluates the efficacy of ON101, a topical macrophage modulator, used alongside systemic antibiotics in managing infected DFUs.

Methods

Retrospective data were collected from 178 patients with Wagner 2 to 4 DFUs who received standard hospitalized care, including pressure relief and adjuvant therapies such as negative pressure wound therapy, dermal regeneration templates, skin grafts/flaps, partial amputations, and silver foam dressings, to reduce ulcer severity. Following this downgrading phase, 88 patients with active infection DFUs (IDSA grade 2/3) receiving systemic antibiotic, 62 patients were treated with ON101 monotherapy and 26 patients continued with adjuvant therapies. The incidence of wound closure in 90 days, 120 days and 150 days and mean healing time were assessed. Amputation incidence in one year post-downgrading phase was also followed up.

Results

The ON101 group significantly increased healing compared to the control group:

	ON101	Control	P-value (Fisher exact test)
At 90 days	62.9% (39/62)	11.5% (3/26)	$p < 0.0001$
At 120 days	75.8% (47/62)	23.1% (6/26)	$p < 0.0001$
At 150 days	85.5% (53/62)	46.2% (12/26)	$p = 0.00013$

Additionally, the mean healing time was significantly shorter in the ON101 group (94.5 days)compared to the control group (144.5 days, $p < 0.0001$). Among the IDSA 2/3 patients, the amputation rate within 1 year after the downgrading process showed that the amputation incidence occurred in ON101 group versus the control was 3.2% (2/62) vs. 19.2% (5/26), with $p = 0.011$.Notably, for those who healed within 120 days, the amputation rates were 0% (0/47) vs. 16.7%(1/6), with $p = 0.0047$.

Discussion

ON101 significantly enhances wound closure and reduces healing time in infected DFUs. At 90 days, ON101achieved a five-fold increase in wound closure compared to the control, demonstrating its potential as an early intervention during infection control to deliver faster and more favorable outcomes. These results highlight the critical role of macrophage modulation in repairing tissue damage and promoting healing within infected wound environments. ON101 offers a compelling therapeutic advantage in managing infected DFUs and reducing the risk of limb loss.

Amputation Rate

	Amputation Rate (1 year after treatment initiation)		P-value (Fisher exact test)
	ON101	Control	-
Infection (IDSA 2/3)	2/62 (3.2%)	5/26 (19.2%)	0.02195
Both infection and PAD	2/57 (3.5%)	5/22 (22.7%)	0.01603

- **Bone Exposure**
- **1.8 tubes 56 days complete healing**

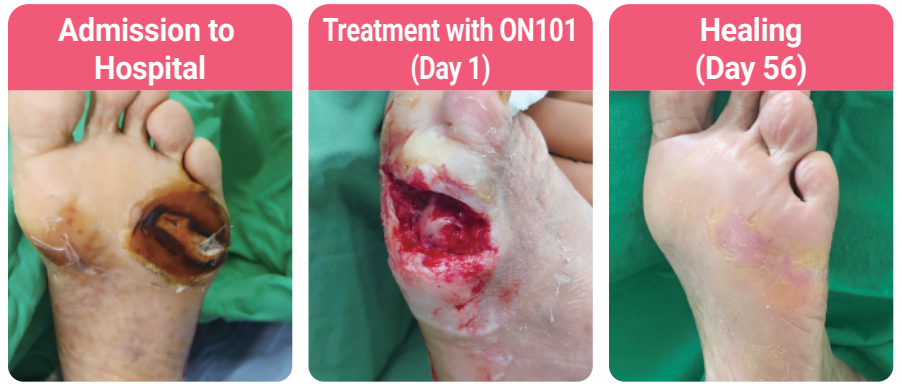
Admission to Hospital
Treatment with ON101 (Day 1)
Healing (Day 56)

Healing Rate ● ON101 ● Current Best Care

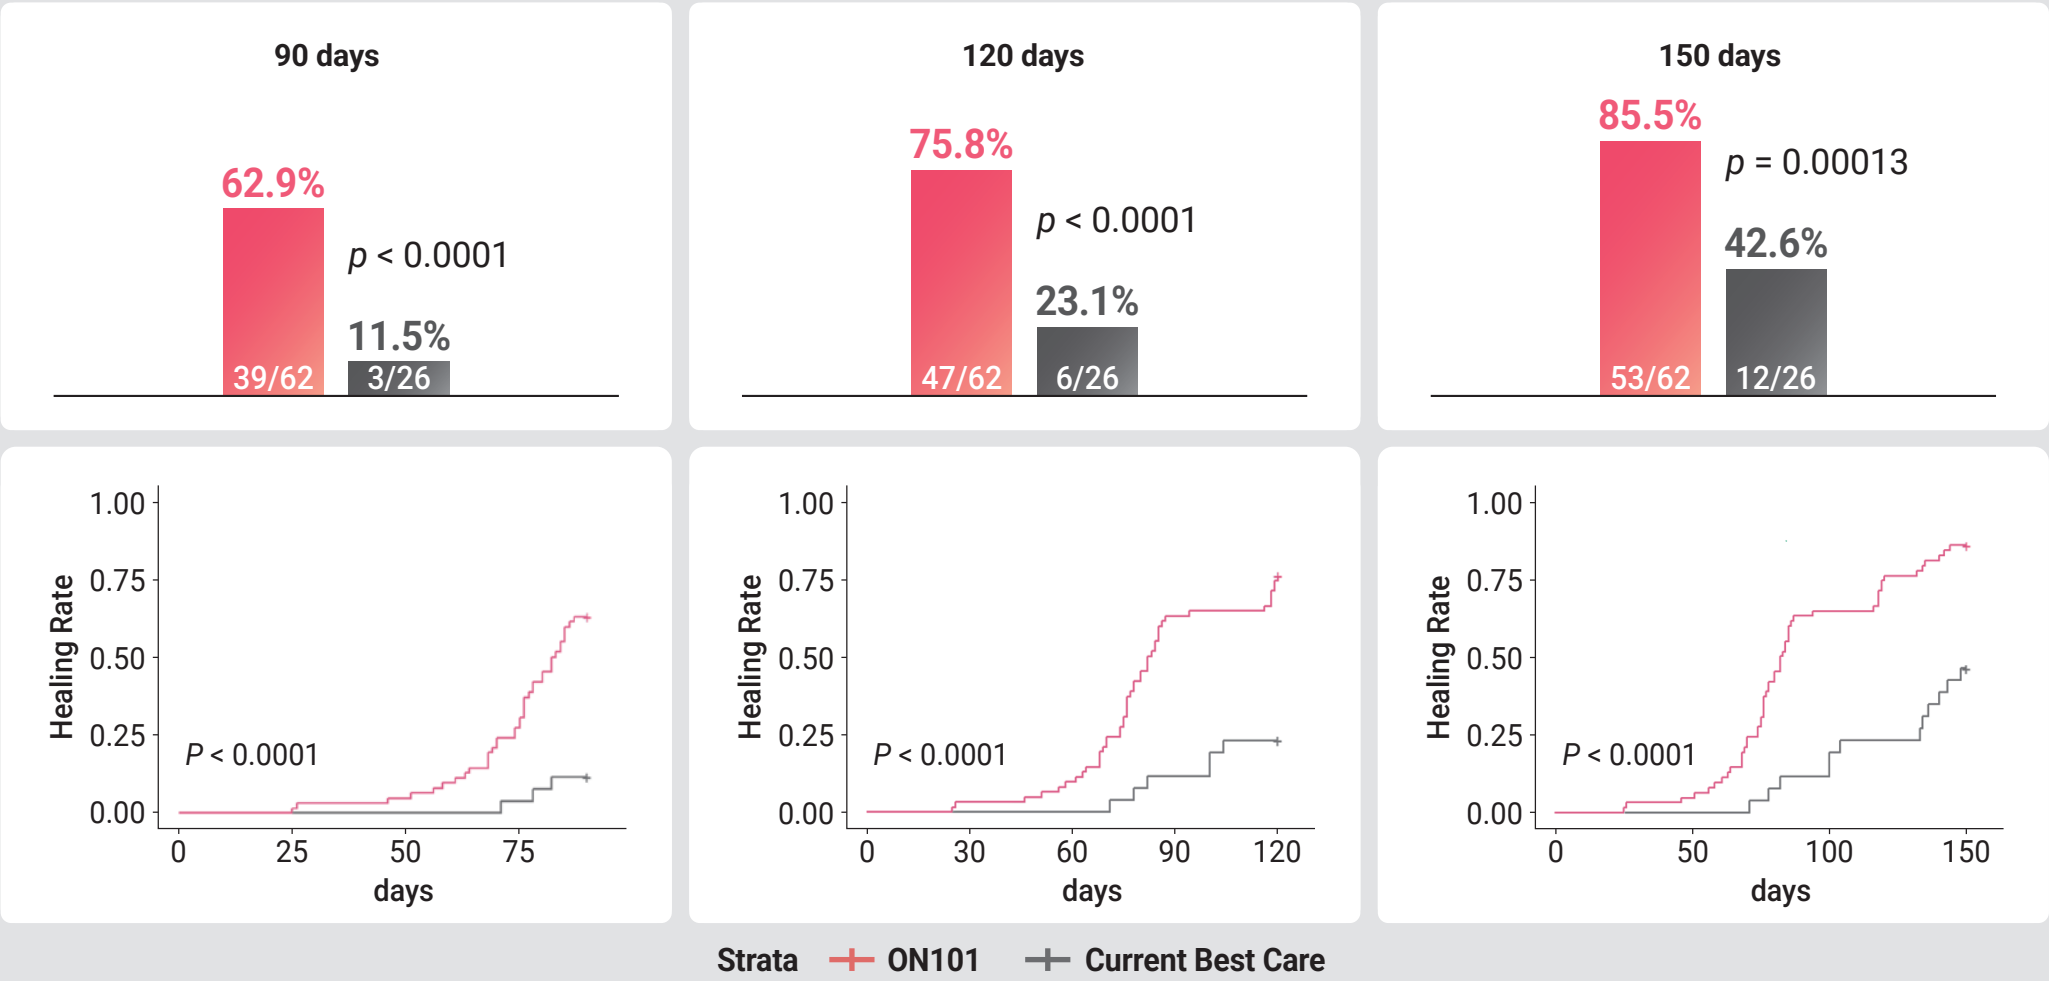
90 days
62.9%
39/62
p < 0.0001
11.5%
3/26
120 days
75.8%
47/62
p < 0.0001
23.1%
6/26
150 days
85.5%
53/62
p = 0.00013
42.6%
12/26
Healing Rate
1.00
0.75
0.50
0.25
0.00
P < 0.0001
days
0
25
50
75
30
60
90
120
100
150
Strata
ON101
Current Best Care

Presented at The Symposium on Advanced Wound Care (SAWC) Spring | Wound Healing Society (WHS), Grapevine, TX, Apr 30 - May 3, 2025